Abstract B25

First-in-human Phase I study to evaluate MP0250, a DARPin® blocking HGF and VEGF, in patients with advanced solid tumors

Jordi Rodon[1], Aurelius Omlin[2], Karin H. Herbschleb[3], Javier Garcia-Corbacho[4], Jan Steiner[5], Ignacio Dolado[6], Christof Zitt[6], Daniel Feurstein[6], Dascha Turner[6], Keith M. Dawson[6], Michael T. Stumpp[6], Patrick Gilboy[6], Andreas Harstrick[6], Analía Azaro[1], Christoph J. Ackermann[2], Mark R. Middleton[3], Richard D. Baird[4]

[1]Vall d'Hebron, Institute of Oncology, Barcelona, Spain; [2]Kantonsspital St Gallen, St Gallen, Switzerland; [3]Oxford University Hospitals NHS Foundation Trust, Churchill Hospital, Oxford, UK; [4]Cambridge University Hospitals NHS Foundation Trust, Addenbrooke's Hospital, Cambridge, UK; [5]Oxford Therapeutics Consulting Ltd, Crowthorne, UK; [6]Molecular Partners, Zurich, Switzerland

Background

DARPins® (designed ankyrin repeat proteins) are small genetically engineered proteins that bind to specific targets with very high affinity (i.e. picomolar; cartoon shown below with target binding sites in color). MP0250 is a multi-DARPin with three specificities, able to simultaneously neutralize the activities of vascular endothelial growth factor (VEGF) and hepatocyte growth factor (HGF) and also to bind to human serum albumin (HSA) to give an increased plasma half-life and potentially enhanced tumor penetration.

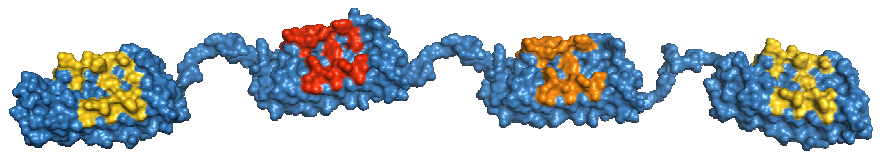

anti-HSA DARPin | anti-VEGF DARPin | anti-HGF DARPin | anti-HSA DARPin

MP0250 is the first systemically delivered DARPin. Like other DARPins, MP0250 can be easily manufactured and is very stable. DARPins are very versatile, can bind to virtually any target, and can be easily formatted in a multi-specific way to block several biological activities.

Preclinical *in vivo* Efficacy

Inhibition of tumor growth in Patient-Derived Xenograft (PDX) models

- MP0250 is a potent inhibitor of tumor growth in multiple patient-derived xenograft models (liver, kidney, stomach, lung, multiple myeloma)
- MP0250 is equal or superior to clinical standard-of-care drugs in all tested models
- MP0250 shows strong additive activity when combined with clinical standard of care
- Potency of MP0250 can be correlated with tumor HGF and VEGF overexpression

PDX renal cancer | PDX gastric cancer

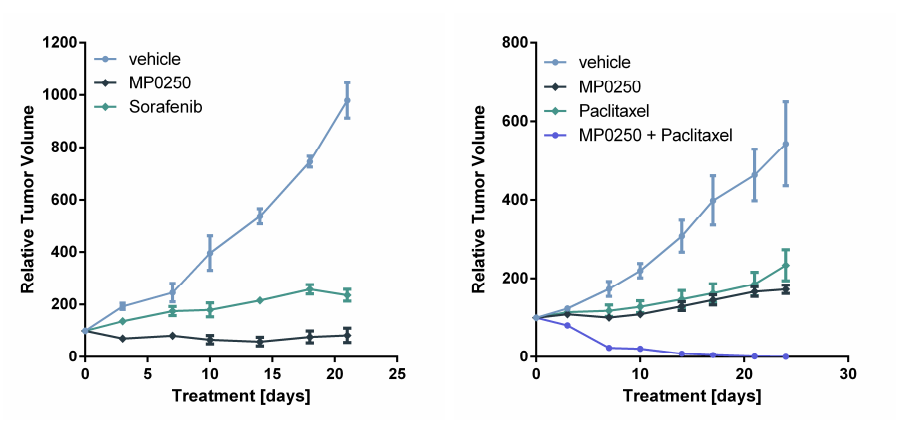
Relative Tumor Volume
Treatment [days]
vehicle
MP0250
Sorafenib
Paclitaxel
MP0250 + Paclitaxel

Antitumor activity of MP0250 in renal and gastric PDX models

MP0250 was dosed at 4 mg/kg 3x weekly (i.v.), sorafenib at 200 mg/kg daily (p.o.) and paclitaxel at 15 mg/kg 1x weekly (i.v.) for 21 days. The combination of MP0250 and paclitaxel was 4 mg/kg MP0250 3x weekly and 15 mg/kg paclitaxel 1x weekly.

Preliminary Study Results

Characteristics of patients enrolled (total and per dose cohort)

Cohort	Dose mg/kg	# Patients	Age (Med., Range)	Male / Female	Tumor types	# Infusions (Med., Range)	# prior Tx (Med., Range)
All	0.5-8	15	63 (20-78)	5/10		5 (1-29)	3 (1-7)
1	0.5	3	70	female	Endometrial sarcoma	8	2
			20	female	Nasopharyngeal carcinoma	29	2
			57	female	Cervical adenocarcinoma	18	3
2	1.5	3	67	male	Colorectal carcinoma	3	7
			60	male	NSCLC	4	4
			62	male	Carcinoma of the tongue	8	1
3	4	6	41	female	Adenocarcinoma of unknown primary	4	1
			67	male	Colorectal carcinoma	1	4
			70	female	Renal cell carcinoma	4	3
			78	female	Breast cancer	4	7
			61	female	Breast cancer	7	1
			63	female	Salivary gland carcinoma	8	0
4	8	3	69	female	NSCLC	8	4
			70	male	Colorectal carcinoma	6	4
			63	female	Ovarian carcinoma	4	2

Patient dosing and efficacy

- Fifteen patients have been enrolled in the first four dose cohorts with MP0250 at 0.5 (n=3), 1.5 (n=3), 4 (n=6), 8 mg/kg (n=3), and the fifth dose cohort with MP0250 at 12 mg/kg every two weeks is currently ongoing
- Stable disease for 12 months (treatment ongoing) has been observed in a patient with a head and neck tumor and for 8 months in a patient with a cervical adenocarcinoma

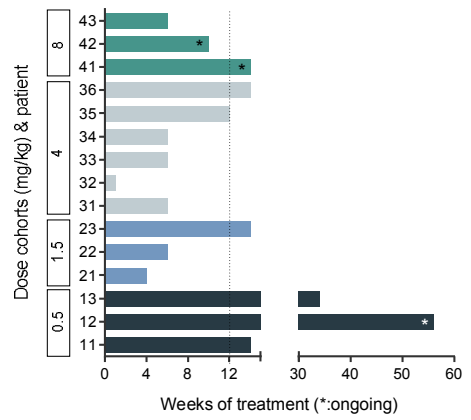
Dose cohorts (mg/kg) & patient
8
4
1.5
0.5
43
42
41
36
35
34
33
32
31
23
22
21
13
12
11
*
0
4
8
12
30
40
50
60
Weeks of treatment (*:ongoing)

Treatment duration was ≥3 months in about 50% of patients with 2 patients exceeding 8 months.

Pharmacokinetics

- Sustained exposure was observed for all patients throughout the treatment periods analyzed, the longest to-date being 12 months
- MP0250 shows a mean half-life of approximately 12 days (range 9-18 days)
- In line with the long half-life of MP0250 slight accumulation was observed (factors 1.3 - 3, based on Cmax, Cmin, and AUC)
- Plasma concentration-time data show low to moderate inter-subject variability (< factor 2-3)

Tolerability and immunogenicity

- MP0250 has been well tolerated and a maximum tolerated dose has not been reached
- The most frequent adverse events (AEs, CTC version 4.03) were transient hypertension (47%), diarrhea (40%), fatigue (40%) and nausea (40%)
- A total of 116 infusions of MP0250 have been administered in 15 patients. Infusions were well tolerated, except for mild nausea and diarrhea occurring in some patients.
- One patient experienced a temporary drop in blood pressure and bradycardia during his first infusion which resolved spontaneously. Even though not classical, an infusion-related reaction could not be excluded in this patient.
- A single dose-limiting toxicity was observed: significant reduction in cardiac ejection fraction after 1st infusion in a patient with multiple cardiac risk factors
- All patients analyzed (15/15) showed sustained exposure, and only 1/8 patients tested showed increasing ADA titers during treatment

List of all adverse events in more than two patients.

Dose cohort (mg/kg)	0.5		1.5		4		8		Total
N	3		3		6		3		15
Severity (NCI-CTC v4.03)	1-2	3	1-2	3	1-2	3*	1-2	3	
Hypertension				1	1	3		2	7
Diarrhea	1		2		2		1		6
Fatigue	1		2		1		2		6
Nausea			1		3		2		6
Cough	2		1		1		1		5
Anorexia	1				2				3
Back pain	1				2				3
Blood bilirubin increased					2		1		3
Vomiting					2		1		3

* Two patients died due to progressive disease; of whom one presented in addition grade 4 hyperglycemia and thrombocytopenia.

Study Protocol

Study summary and objectives

This is a phase I first-in-human multi-center, open-label, repeated-dose, dose-escalation (3+3 design) study to assess safety, tolerability, pharmacokinetics and immunogenicity of i.v. MP0250 in patients with advanced solid tumors.

Primary objectives:

- Evaluate the safety and tolerability of MP0250
- Determine the Maximum Tolerated Dose (MTD), Recommended Biological Dose (RBD) and Dose Limiting Toxicities (DLTs) of MP0250
- Characterize the pharmacokinetics of MP0250

Secondary objectives:

- Characterize the immunogenicity of MP0250
- Make a preliminary assessment of biomarkers and genetic markers
- Evaluate the anti-tumor activity of MP0250

Patients and methods:

Key inclusion criteria: Histologically confirmed advanced or metastatic solid tumor refractory to ≥ 1 prior regimen of standard treatment or for which no curative therapy is available; Progressive or stable disease documented radiologically in the 4 weeks prior to screening; Presence of a measurable tumor or a tumor evaluable per RECIST v1.1; Serum albumin concentration ≥ 30 g/L.

Key exclusion criteria:
Hematological malignancies or other secondary malignancy; Known untreated or symptomatic brain metastases; Predominantly squamous non-small cell lung carcinoma; Anti-tumor treatment less than 4 weeks prior to the first infusion of MP0250, such as chemotherapy, experimental or targeted therapy, biologics, hormonal therapy and radiotherapy; Proteinuria; Uncontrolled hypertension.

Study treatment:

- Enrollment into five dose cohorts: 0.5, 1.5, 4, 8, 12 mg/kg
- Up to 12 intravenous infusions of MP0250 without pre-treatment: every two weeks administered over 3 hours
- Treatment until disease progression, DLT or other reasons for withdrawal. Patients are allowed to continue treatment >12 infusions in case of benefit

Conclusion

- This is the first demonstration of the systemic application of a DARPin in patients and shows the feasibility of the approach in oncology
- MP0250 is a first-in-class dual inhibitor of HGF and VEGF with potential to treat patients with various solid tumors
- MP0250 is well tolerated at doses ranging from 0.5 to 8 mg/kg given every two weeks as i.v. infusion, with two patients having stable disease for >12 months and 8 months, respectively
- MP0250 shows a long half-life of around 12 days, giving the potential of dosing every three weeks and beyond
- Repeated dosing led to sustained exposure throughout the treatment periods analyzed, the longest to-date being 12 months
- These preliminary results provide the basis of the phase II strategy for MP0250 and the overall strategy for the DARPin® technology platform

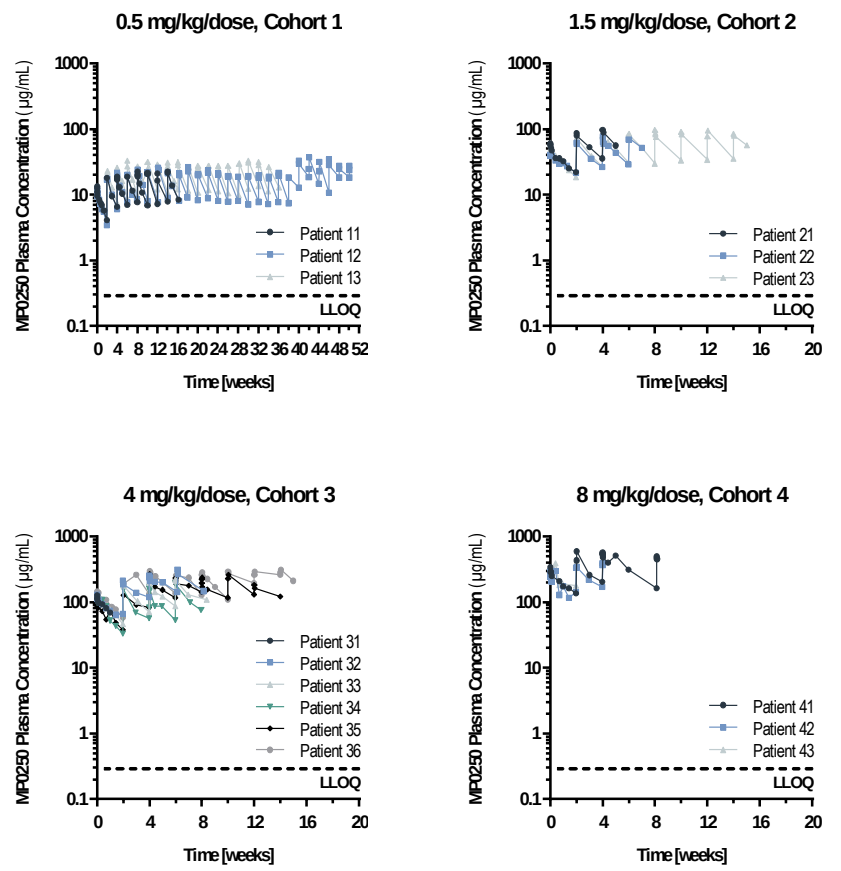
0.5 mg/kg/dose, Cohort 1
1.5 mg/kg/dose, Cohort 2
4 mg/kg/dose, Cohort 3
8 mg/kg/dose, Cohort 4
MP0250 Plasma Concentration (µg/mL)
Time [weeks]
LLOQ
Patient 11
Patient 12
Patient 13
Patient 21
Patient 22
Patient 23
Patient 31
Patient 32
Patient 33
Patient 34
Patient 35
Patient 36
Patient 41
Patient 42
Patient 43

Concentration-time data of MP0250 in cancer patients

Plasma concentration-time data of MP0250 in individual cancer patients infused over 3hrs once every two weeks at dose levels of 0.5, 1.5, 4, and 8 mg/kg/dose. Time: Time at which samples were actually taken post end of first infusion. LLOQ: Lower limit of quantification. An ELISA is used to determine MP0250 concentration in patient plasma samples. Human HGF is coated onto microplates to capture MP0250. A primary anti-DARPin antibody and a secondary antibody/HRP conjugate is used for detection.

Molecular Partners AG, Wagistrasse 14, 8952 Zurich-Schlieren, Switzerland – www.molecularpartners.com – info@molecularpartners.com – phone: +41 44 755 77 00